Synthetic matrix-mimetic polypeptide constructs enhance attachment of mesenchymal cells to diverse scaffold surfaces

Áron Szepesi[1], Anna Szigeti[1], Péter Tátrai[2,3], Ildikó Szabó[4], Gábor Mező[4], Katalin Német[1,2]

[1] Creative Cell Ltd., Budapest, Hungary; [2] Dept. of Experimental Gene Therapy, National Blood Transfusion Service, Budapest, Hungary; [3] Dept. of Biochemistry and Molecular Biology, University of Debrecen, Debrecen, Hungary; [4] Research Group of Peptide Chemistry, Hungarian Academy of Sciences, Eötvös Loránd University, Budapest, Hungary

Introduction. For both bone tissue regeneration and implantation, efficiency of cell attachment to the scaffold or implant surface is critical to success. However, several widely used surgical and implant materials have limited ability to promote cell adhesion. Failure of cells, either engrafted or host, to adhere to the surface may impede regeneration or lead to implant loosening.

Aims. We developed novel RGD-containing synthetic polypeptide conjugates, and tested their ability to improve attachment of human adipose tissue-derived mesenchymal cells (MSCs) to diverse surfaces such as plastic, titanium, bovine bone mineral, and a copolymeric surgical mesh. Cell adhesion experiments were carried out under restrictive, serum-free conditions.

Materials and methods

Polypeptide conjugates. To yield SAK-cRGD, the RGD-containing cyclic pentapeptide cRGDfC (shortly, cRGD) was covalently linked to a backbone termed SAK, consisting of poly-L-lysine with serine-oligo-DL-alanine branches. To yield SAK-cRGD-OPN, the osteopontin-derived linear peptide CGRGDSVVYGLR was also linked to SAK in addition to cRGD. Surfaces were coated by simple adsorption of the conjugates.

Scaffold materials. Titanium mesh with a wire diameter of 228.6 µm was obtained from Unique Wire Weaving (Hillside, NJ, USA), and cut into 3x3 mm pieces. Bio-Oss® bovine bone mineral (granule size: 0.25-1 mm) was from Geistlich Biomaterials (Wolhusen, Switzerland). TIGR™ surgical mesh was from Novus Scientific (Uppsala, Sweden).

Human adipose tissue-derived MSCs. All MSCs used in the experiments were isolated from a single 30-year-old female donor by standard protocol (collagenase digestion and selection for plastic adherence), with written consent from the patient and permission from the Ethical Committee. An aliquot of cells at passage 5 was transduced with eGFP-encoding lentivirus at a multiplicity of infection of cca. 2, and sorted for GFP-positivity by FACS.

Determination of cell number. Cell numbers were estimated by Resazurin conversion assay. Cells were incubated with Resazurin dye, fluorescence was measured at 579 nm, and cell number was calculated using a calibration curve.

Further methods are referred to in the Results *section.*

Results

Figure 1. Dose response curve of seeding efficiency. Similar to fibronectin (FN), both SAK-cRGD and SAK-cRGD-OPN exerted a concentration-dependent positive effect on the adhesion of MSCs to plastic. The adhesion-enhancing effect of the conjugates was half-maximal at or below 0.1 µg/ml, and was well in saturation at 10 µg/ml. The maximal increase in adhered cell number (at 10µg/ml) was 69±13% and 58±15% for SAK-cRGD and SAK-cRGD-OPN, respectively. Unconjugated cRGD was inefficient, while SAK alone inhibited adhesion in a concentration-dependent manner.

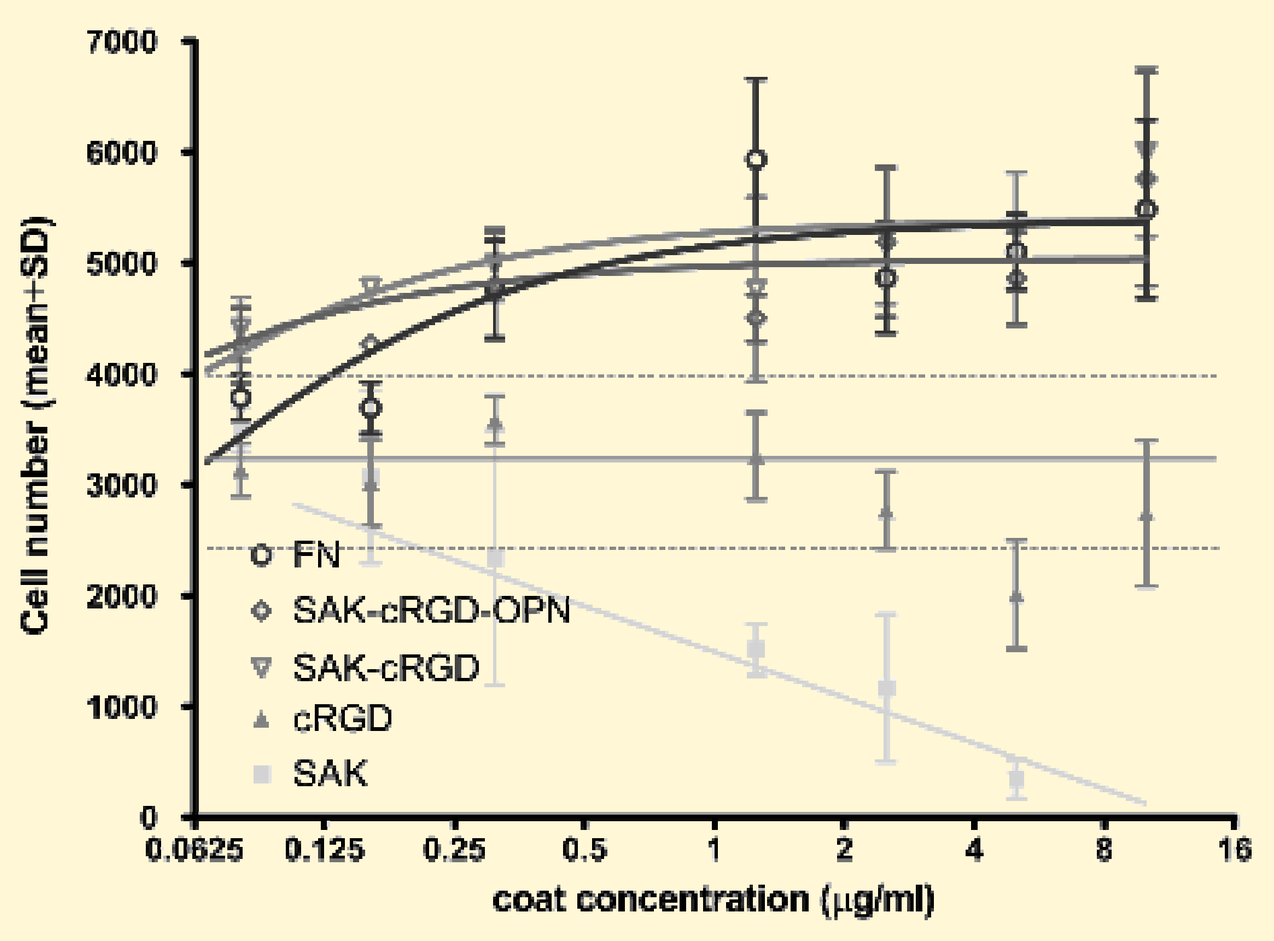
Cell number (mean+SD)
coat concentration (µg/ml)
FN
SAK-cRGD-OPN
SAK-cRGD
cRGD
SAK

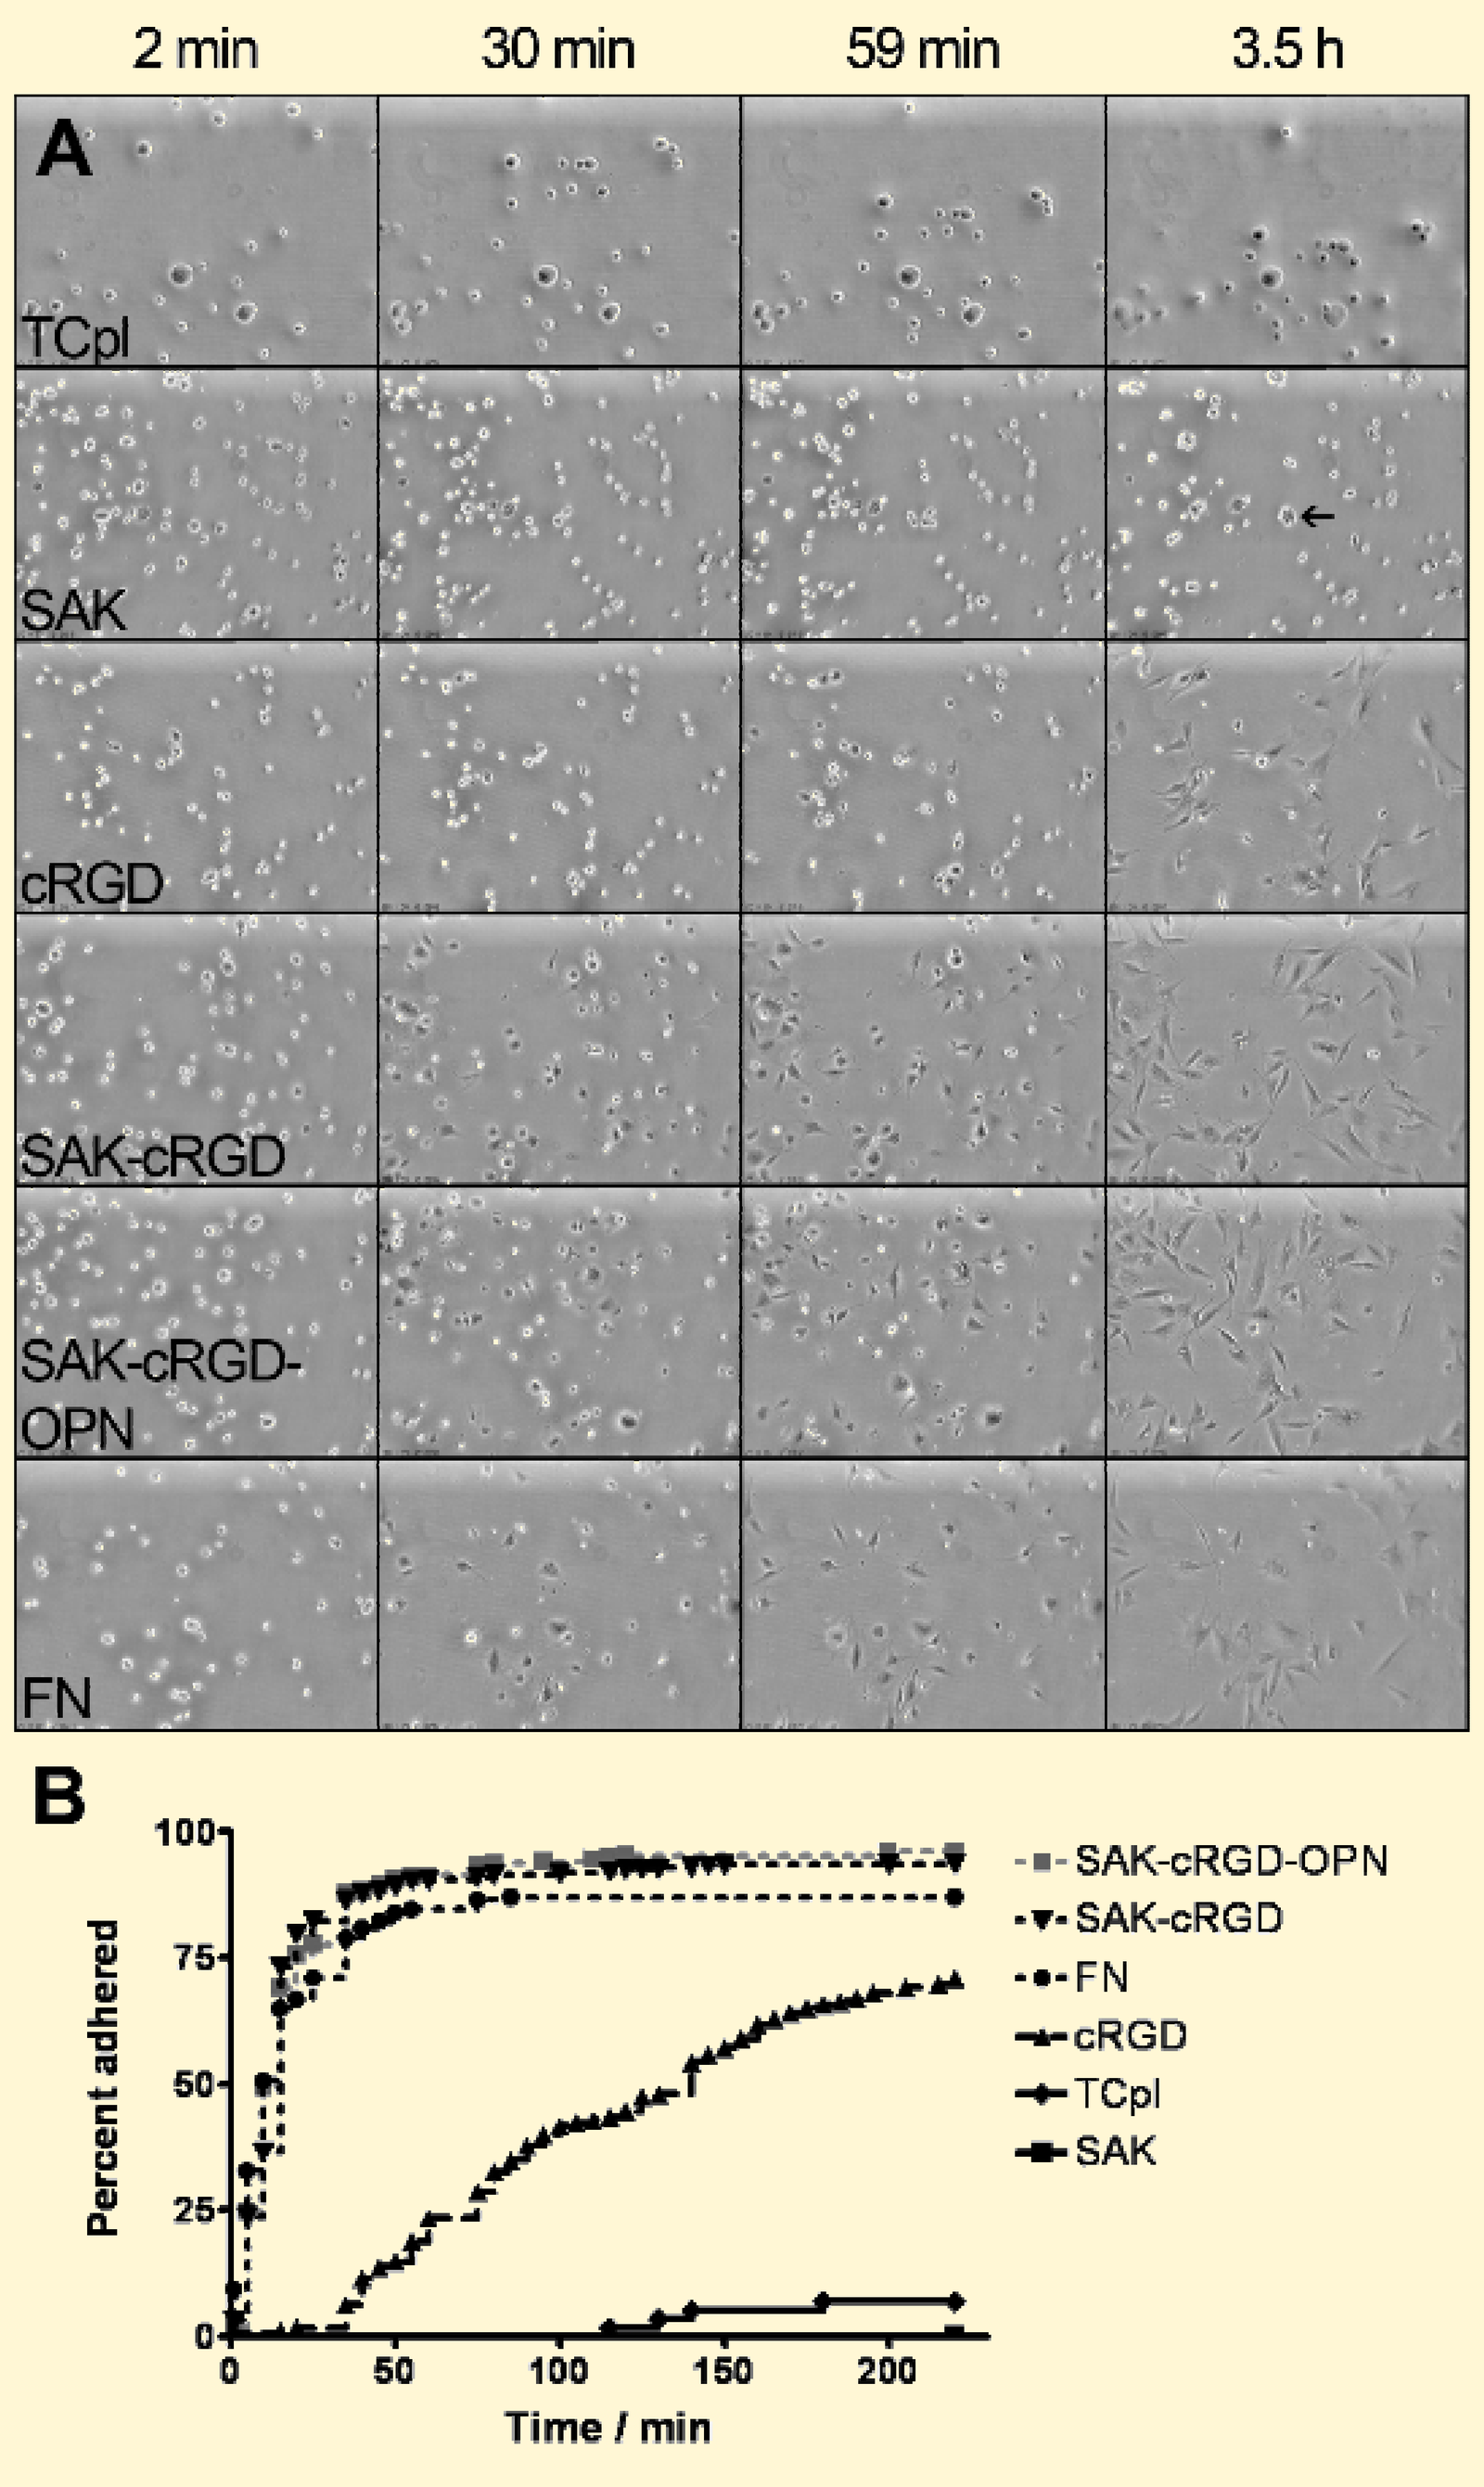
2 min
30 min
59 min
3.5 h
A
TCpl
SAK
cRGD
SAK-cRGD
SAK-cRGD-OPN
FN
B
Percent adhered
Time / min
SAK-cRGD-OPN
SAK-cRGD
FN
cRGD
TCpl
SAK

Figure 2. Time course of adhesion. (A) Phase contrast micrographs of cells seeded on untreated (TCpl) vs. treated plastic, taken at various time points. (B) The percent of adhered cells plotted against time. On plastic treated with the conjugates, 15 min was required for the anchoring of 50% of cells (FN: 10 min, cRGD: 140 min). No adhered cells were observed on TCpl and SAK; on the latter, cells rather tended to form clusters (arrow).

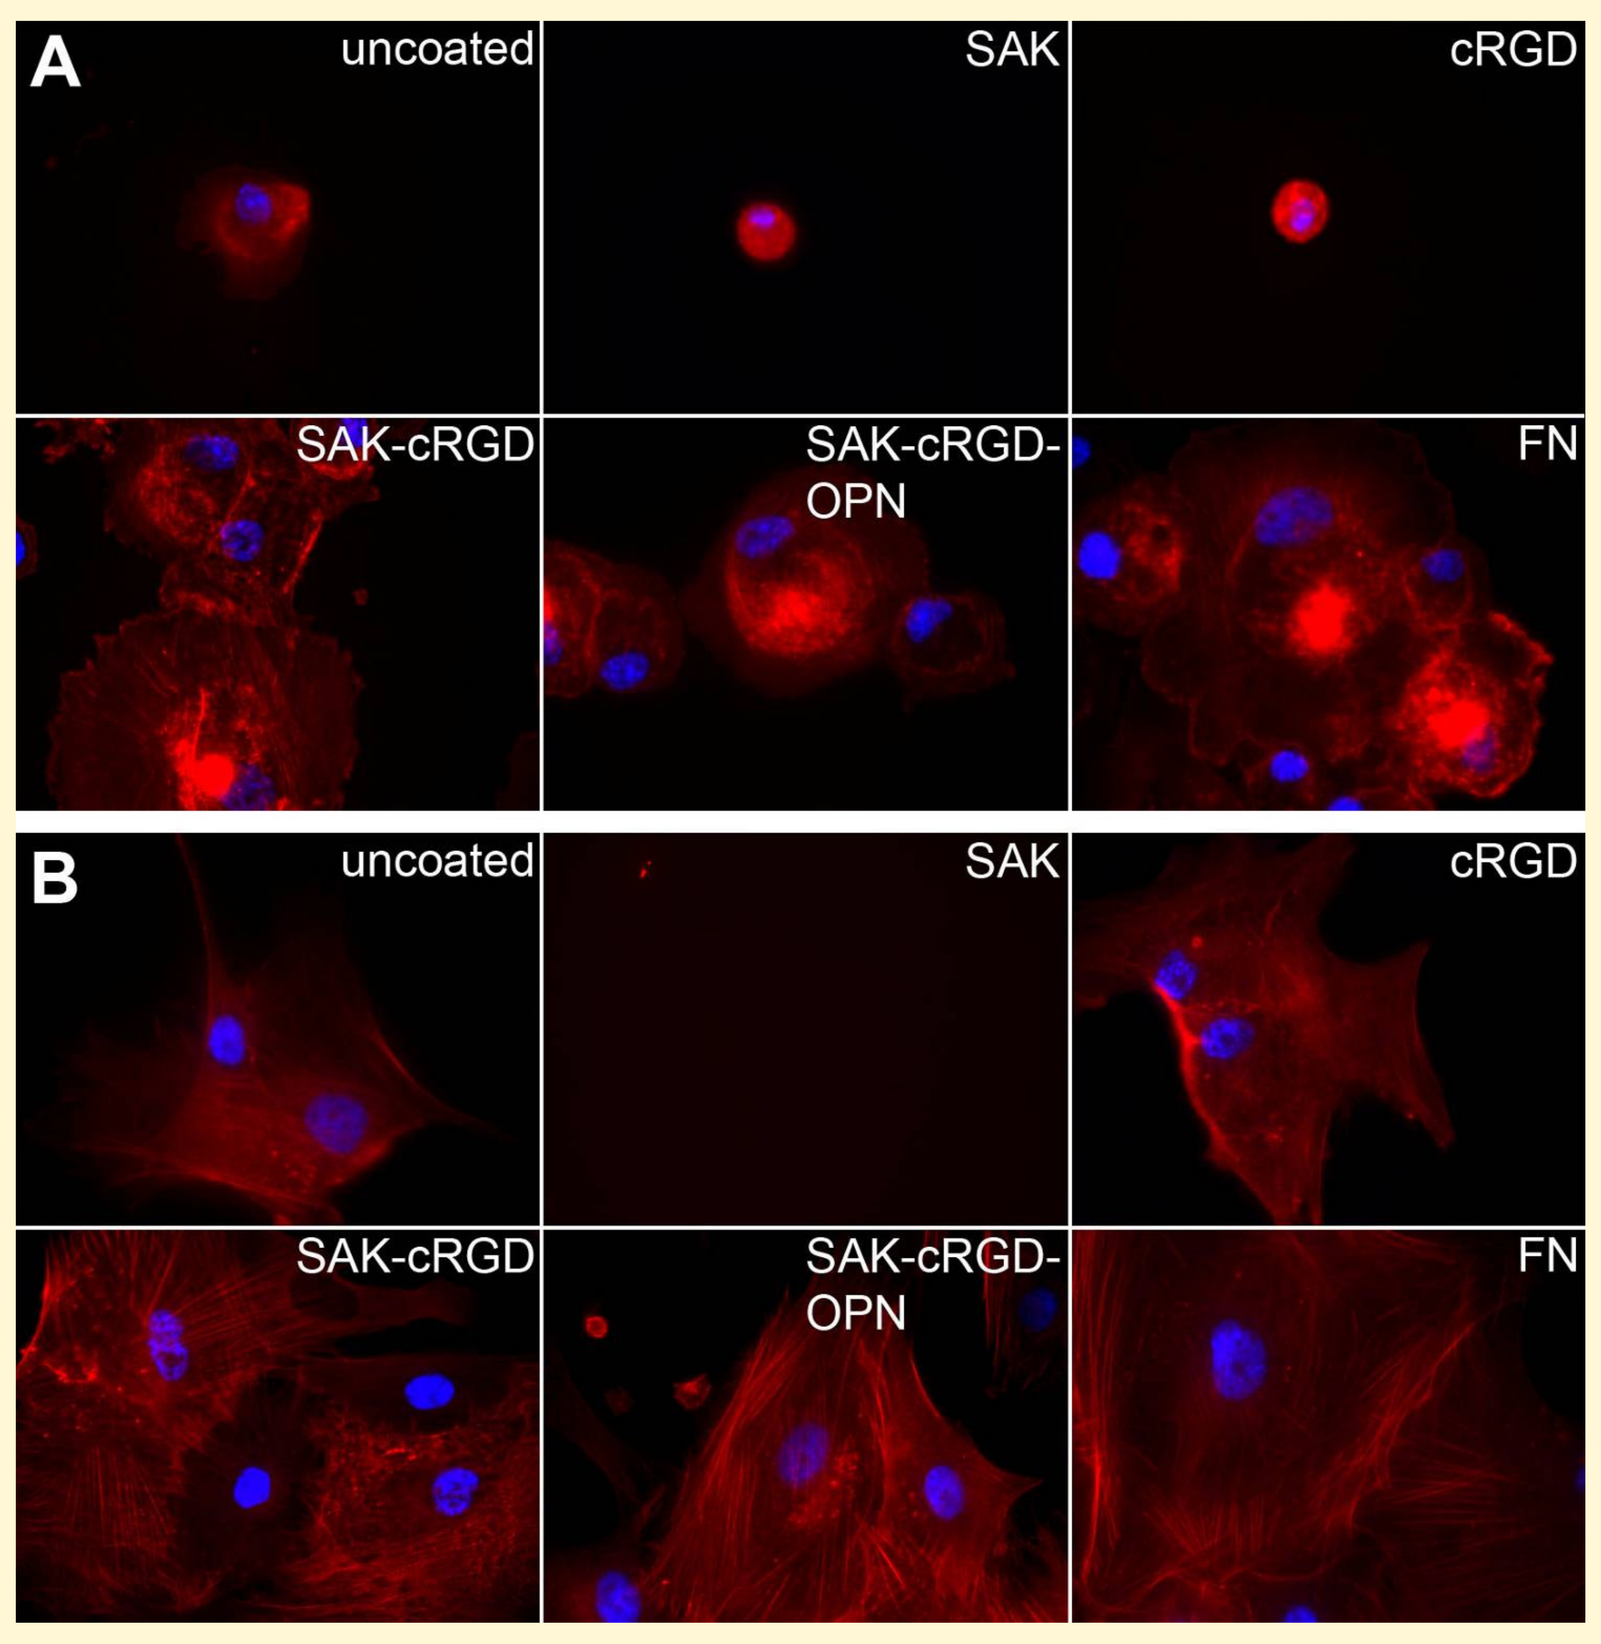
A
uncoated
SAK
cRGD
SAK-cRGD
SAK-cRGD-OPN
FN
B

Figure 3. Cytoskeletal organization of attached cells. MSCs seeded on uncoated vs. coated plastic were stained with TRITC-labeled phalloidin after (A) 30 min and (B) 4 h of attachment. Stress fiber formation on conjugate-coated plastic started at 30 min and was extensive after 4 h.

Figure 4. Adhesion to uncoated vs. coated titanium and Bio-Oss. GFP-expressing MSCs were seeded on (A) titanium or (C) Bio-Oss for 4 h, unattached cells were aspirated, the constructs were incubated for additional 1 day in serum-free medium, and photographed. Alternatively, non-fluorescent cells were seeded for 30 min or 4 h, and quantified after 1 day (B, titanium and D, Bio-Oss). On titanium, seeding efficiency was significantly enhanced by the conjugates only with prolonged seeding, whereas on Bio-Oss, significant enhancement was observed after as few as 30 min.

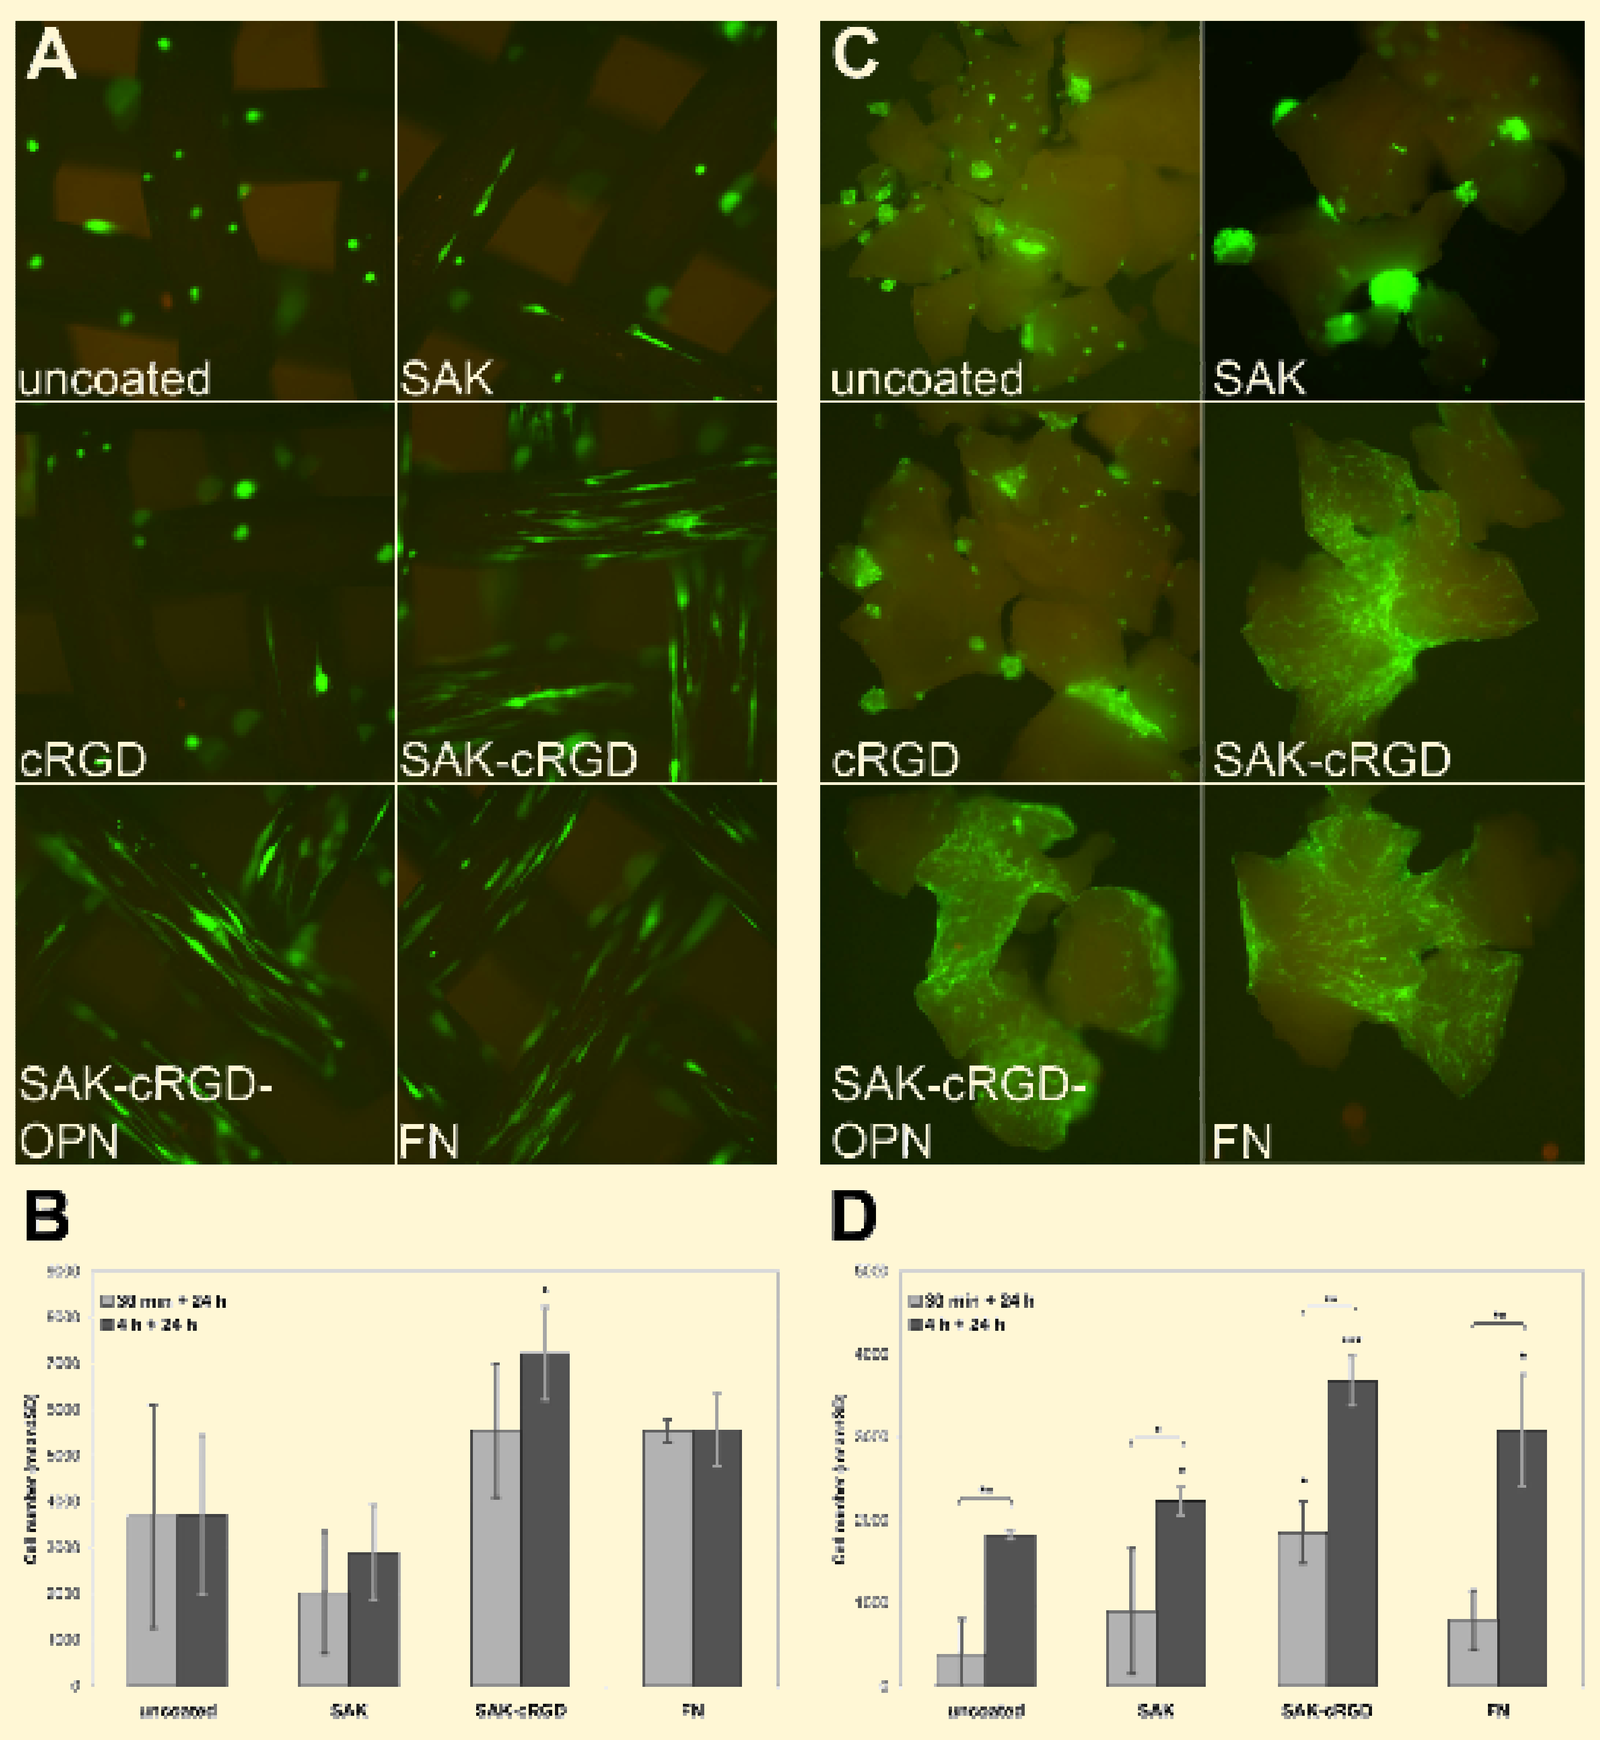
A
C
uncoated
SAK
cRGD
SAK-cRGD
SAK-cRGD-OPN
FN
B
D

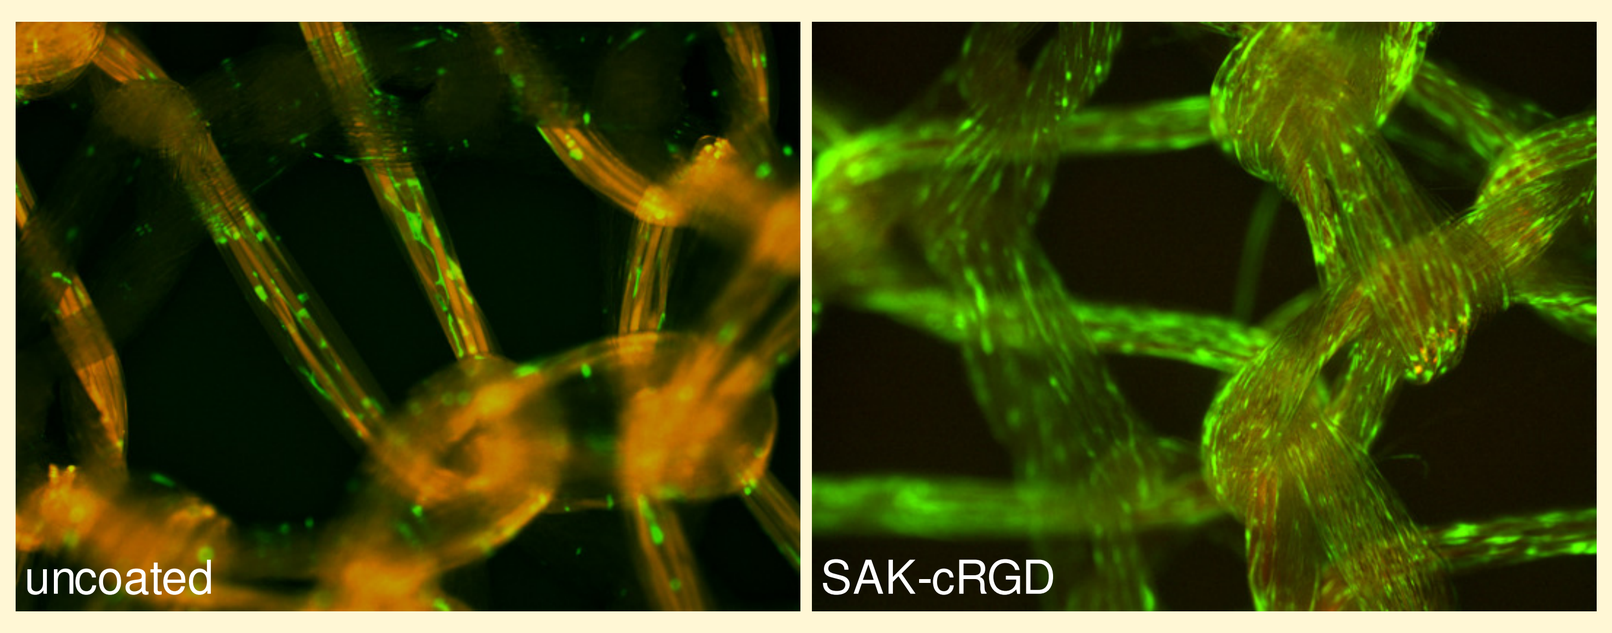
uncoated
SAK-cRGD

Figure 5. Adhesion to uncoated vs. coated TIGR mesh. Following a 4-h seeding and a 1-day incubation period, GFP-MSCs attached to uncoated and SAK-cRGD-coated TIGR mesh were photographed.

Figure 6. Morphology of cells attached to uncoated vs. coated Bio-Oss by scanning electron microscopy. The very few cells attached to uncoated and cRGD-coated Bio-Oss remained round or incompletely spread. On SAK coating, cells clumped together and made little contact with the surface. On the conjugate-coated surfaces, however, cells were neatly spread and developed numerous extensions.

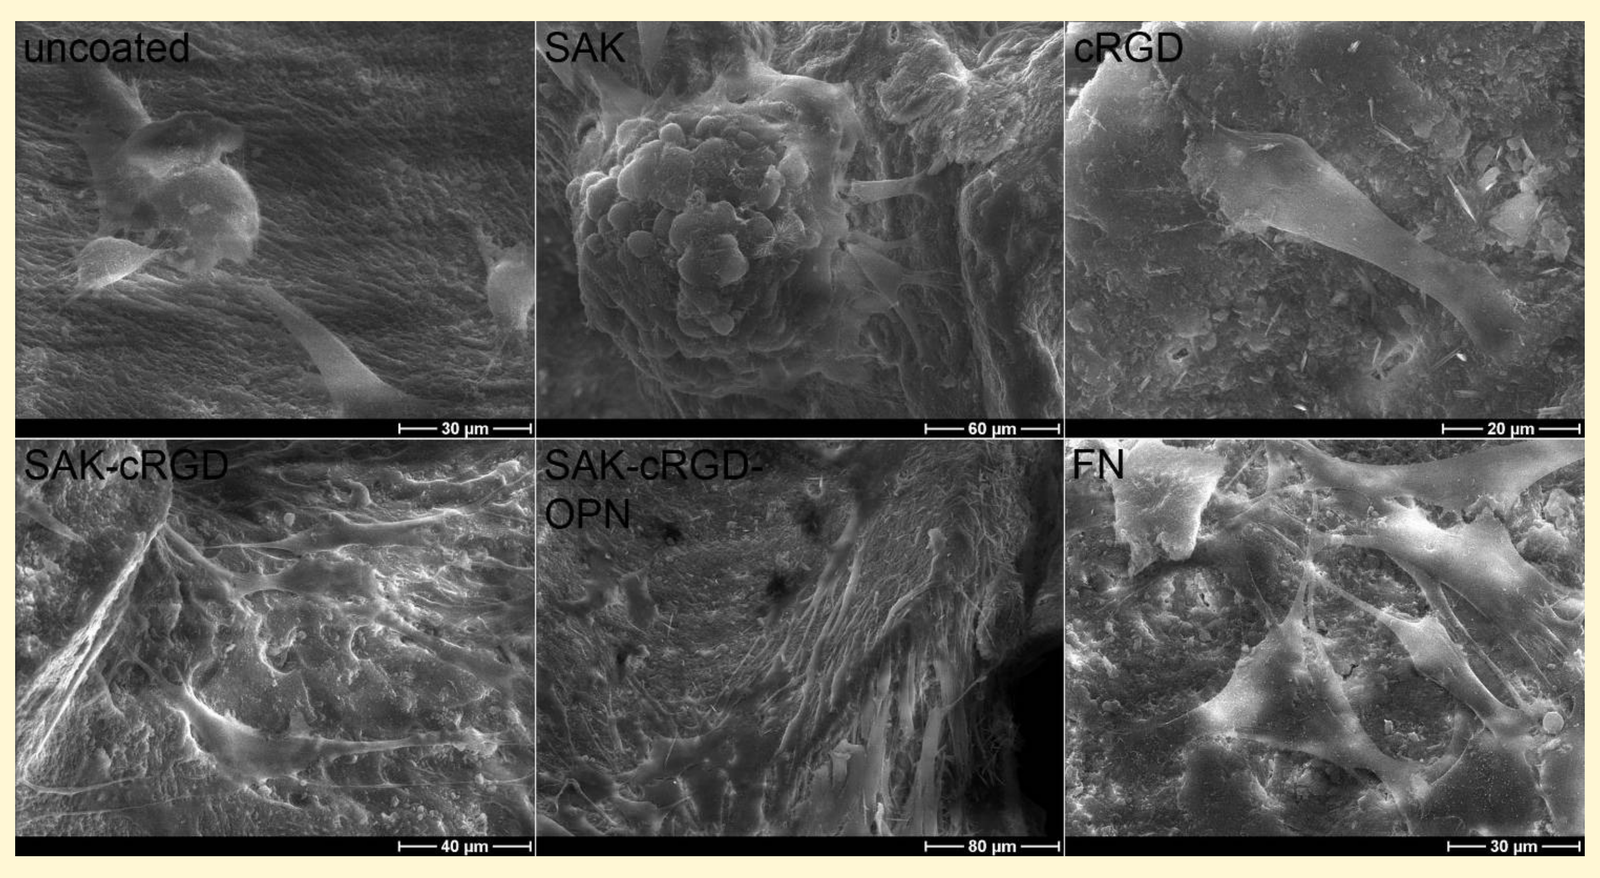
uncoated
SAK
cRGD
SAK-cRGD
SAK-cRGD-OPN
FN

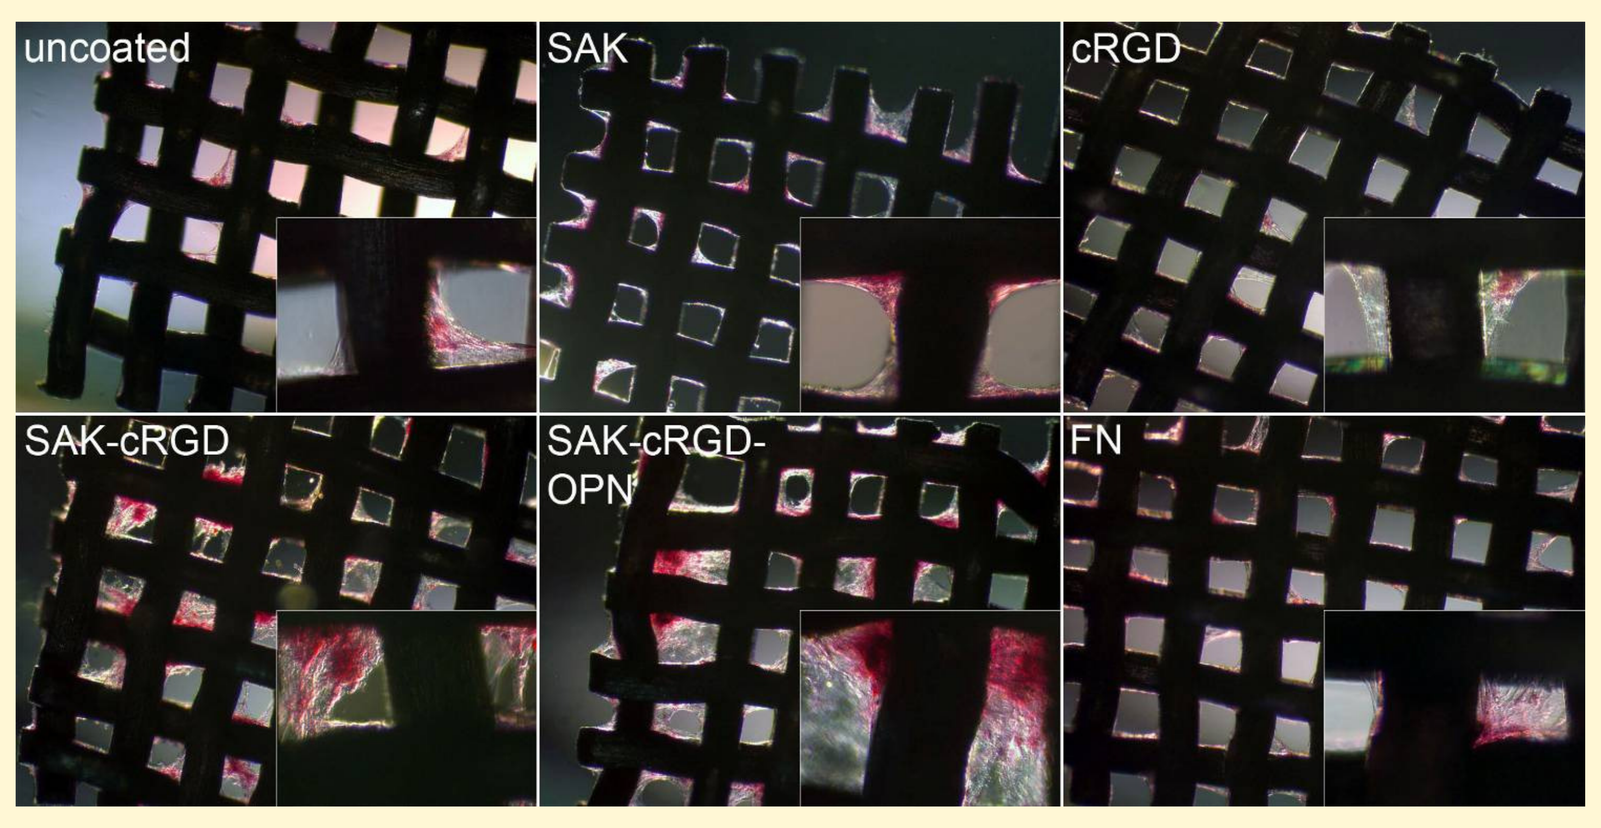
uncoated
SAK
cRGD
SAK-cRGD
SAK-cRGD-OPN
FN

Figure 7. Calcification of MSCs on uncoated vs. coated titanium meshes. MSCs seeded without serum on uncoated and coated titanium were subjected to bone differentiation for 28 days in serum-containing osteoinductive medium, and stained with Alizarin red. Due to the higher initial cell density, more massive calcification was achieved on the conjugate-coated meshes.

Conclusions

Our observations confirmed that the polypeptide conjugates SAK-cRGD and SAK-cRGD-OPN increased the affinity of surfaces to cells with an efficiency comparable to that of fibronectin. Human adipose tissue-derived MSCs rapidly anchored to the coated surfaces and remained firmly attached in the absence of serum. The coatings also supported long-term culture necessary for *in vitro* differentiation.

As these conjugates can be manufactured in a reproducible and cost-efficient manner, lyophilized and stored indefinitely, reconstituted easily, applied to a surfaces by simple absorption, and remain unchanged on the surfaces under normal conditions, they may provide a reasonable alternative to recombinant protein-based surface treatment.

Institutions:

Funding:
TÁMOP-4.2.1-IKUT (NFÜ)
BIO_SURF (NKTH)